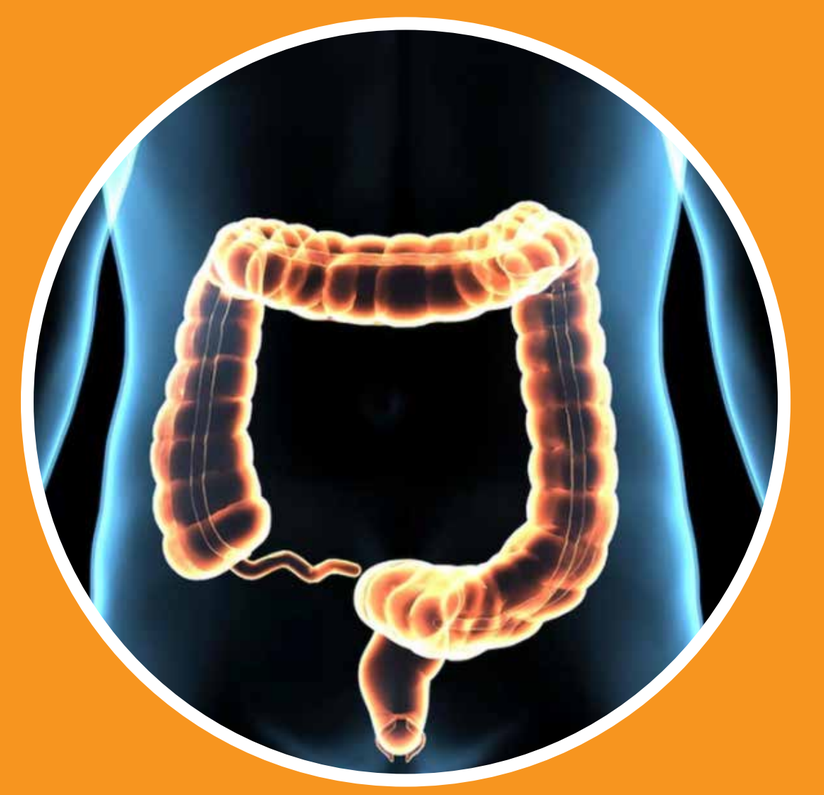

Folate gene prediction of treatment response to 5-FU and leucovorin in metastasizing colorectal cancer

B Gustavsson[1], E Odin[1], P Saksena[2], H Wedel[3], A Edsjö[4], G Carlsson[1], H Taflin[1], E Bexe Lindskog[1], K Ganlöv[5], A Vedin[5], M Åström[6], Y Wettergren[1]

[1]Department of Surgery, Sahlgrenska Academy at University of Gothenburg, Gothenburg, Sweden; [2]Department of Pathology, Sahlgrenska University Hospital, Gothenburg, Sweden; [3]BioStatisticsConsultants, Gothenburg, Sweden; [4]Department of Clinical Genetics and Pathology, Regional Laboratories Region Skåne, Lund, Sweden; [5]Isofol Medical AB, Arvid Wallgrens backe 20, Göteborg, Sweden ; [6]StatCons, Malmö, Sweden

UNIVERSITY OF GOTHENBURG

INTRODUCTION

The combination 5-fluorouracil (5-FU) and leucovorin (LV) has long been central in chemotherapy for colorectal cancer (CRC). Modulation of 5-FU with LV increases the treatment response from 11% to about 21%[1]. LV requires metabolic activation to the co-factor [6R]-5,10-methylenetetrahydrofolate. We have previously found a positive correlation between survival and expression of folate pathway genes in stage III CRC treated with adjuvant FLV (5-FU + LV)[2]. Low expression of folate-related genes may lead to poor response to FLV since poor transport and metabolization of LV might yield insufficient levels of co-factor leading to weak inhibition of the target enzyme thymidylate synthase (TYMS).

The aim of the present study was to relate progression-free survival (PFS) with the expression of defined folate-relevant genes in tumor biopsies of patients with metastasizing CRC (mCRC).

PATIENTS AND METHODS

A total of 294 patients with mCRC receiving first line treatment with bolus FLV, FLV + oxaliplatin (FLOX) or FLV + irinotecan (FLIRI) at the Sahlgrenska University Hospital, Gothenburg, Sweden, during 1995-2012 were included in the study. Patient and tumor characteristics are presented in **Table 1.** All tumors were classified according to the Tumor-Node-Metastasis (TNM) staging system when the patients were first diagnosed (index biopsy occasion). There were 151 patients with Stage 4 (mCRC) already at the index biopsy occasion (synchronous mCRC) and 143 patients with a previously known diagnosis of CRC (Stages 1-3) later manifesting mCRC (metachronous mCRC) who initiated the start of treatment and the present follow-up. The patients were followed after start of treatment for three years or until disease progression or death. PFS was calculated from the date of treatment start to the last follow up, or to the date of progression or death.

Total RNA was isolated. cDNA was synthesized using reverse transcription. The relative expression of 10 genes putatively involved in LV transport, polyglutamation, and metabolism (**Table 2**) was quantified using real time PCR (qPCR).

The Cox proportional hazards regression model was used to examine the relationship between clinical variables, (age, sex, tumor differentiation, ECOG grade, therapy (FLV, FLOX, FLIRI)) chosen tumor gene expression levels and PFS followed by correction for multiple testing. Kaplan-Meier curves for PFS were constructed using tertiles as strata (p value=log-rank test for trend). All the tests were two-tailed and conducted at a significance level of 0.05.

RESULTS

Among metachronous mCRC patients, only treatment and ECOG class, but no gene variable, had significant correlations with PFS (Table 3). Among synchronous patients there were several significant positive correlations between PFS and expression of several folate relevant genes. Inclusion of the clinical covariates age, sex and tumor differentiation in the analysis did not significantly contribute to the multivariate prognostic model and the impact of ECOG and treatment only influenced the HR and p-values in a minor way.

Among synchronous patients, after adjustment for multiple testing in the Cox analysis of all analyzed genes, only ABCC3 remained significant ($p<0.0002$). The ABCC3 expression levels were divided into tertiles; the low, intermediate and high levels. Patients in the intermediate and low tertiles had near identical outcomes and, when combined, had a median PFS of 6.5 months, compared to 10.1 months among patients in the high expression group ($p<0.002$; Fig. 1).

The ABCC3 protein is involved in outward transport of folates, and has preference for 10-formyltetrahydrofolate, which inhibits the first conversion step of LV. High expression of ABCC3 may therefore decrease intracellular levels of 10-formyltetrahydrofolate leading to a high conversion rate of LV to co-factor, and enhanced inhibition of TYMS.

.

Table 1. Patient characteristics

	Metachronous mCRC	Synchronous mCRC
N	143	151
Mean Age	67,2	63,4
Std. dev.	9,7	10,6
Median Age	69,0	64,0
Men	83	75
Women	60	76
ECOG grade		
0	85	116
1-2	58	35
Tumor stage at primary surgery		
1-3	143	0
4	0	151
Treatment		
FLV	52	62
FLOX	49	53
FLIRI	42	36

Table 3. Univariate Cox proportional hazard model using PFS as dependent variable. Gene expression and clinical covariates were included one at a time (HR = Hazard Ratio).

Independent variables	Metachronous mCRC (N=143)		Synchronous mCRC (N=151)	
	HR	P-value	HR	P-value
Genes				
ABCC1	1.0417	0.7060	1.3457	0.0150
ABCC3	0.9345	0.4800	1.3781	0.0000
FPGS	0.9646	0.5390	1.1091	0.0160
GGH	1.0747	0.2130	1.0407	0.4650
MTHFD1L	1.0702	0.5540	1.1260	0.3110
MTHFD2	0.9425	0.4410	0.7823	0.0130
SHMT1	1.1372	0.1540	1.1777	0.0190
SHMT2	1.0770	0.5290	1.1555	0.2580
SLC19A1	1.0556	0.5250	1.2994	0.0010
SLC46A1	0.9335	0.2900	1.1938	0.0050
Clinical covariates				
ECOG	2.2311	0.0000	2.5594	0.0000
Treatment	0.7110	0.0020	0.7982	0.0280

Table 2. Analyzed genes with putative impact on LV metabolism plus two reference genes

Gene category	Gene	Gene name	Assay ID
Folate Transport	ABCC1/MRP1	ATP-binding cassette, subfamily C (CFTR/MRP), member 1	Hs00219905_m1
	ABCC3/MRP3	ATP-binding cassette, subfamily C (CFTR/MRP), member 3	Hs00358656_m1
	SLC19A1/RFC-1	Solute carrier family 19 (folate transporter), member 1	Hs00953344_m1
	SLC46A/PCFT	Solute carrier family 46 (folate transporter), member 1	Hs00611081_m1
Folate polyglutamation	FPGS	Folylpolyglutamate synthase	Hs00191956_m1
	GGH	Gamma-glutamyl hydrolase (conjugase, folylpolygammaglutamyl hydrolase)	Hs00914163_m1
Folate metabolism	MTHFD1L	Methylenetetrahydrofolate dehydrogenase (NADP+ dependent) 1-like	Hs00383616_m1
	MTHFD2	Methylenetetrahydrofolate dehydrogenase (NADP+ dependent) 2	Hs00759197_s1
	SHMT1	Serine hydroxymethyltransferase 1 (soluble)	Hs00541038_m1
	SHMT2	Serine hydroxymethyltransferase 2 (mitochondrial)	Hs00193658_m1
Reference genes	ACTB	Beta-actin	Hs01060665_g1
	PPIA	Peptidylprolyl isomerase A	Hs99999904_m1

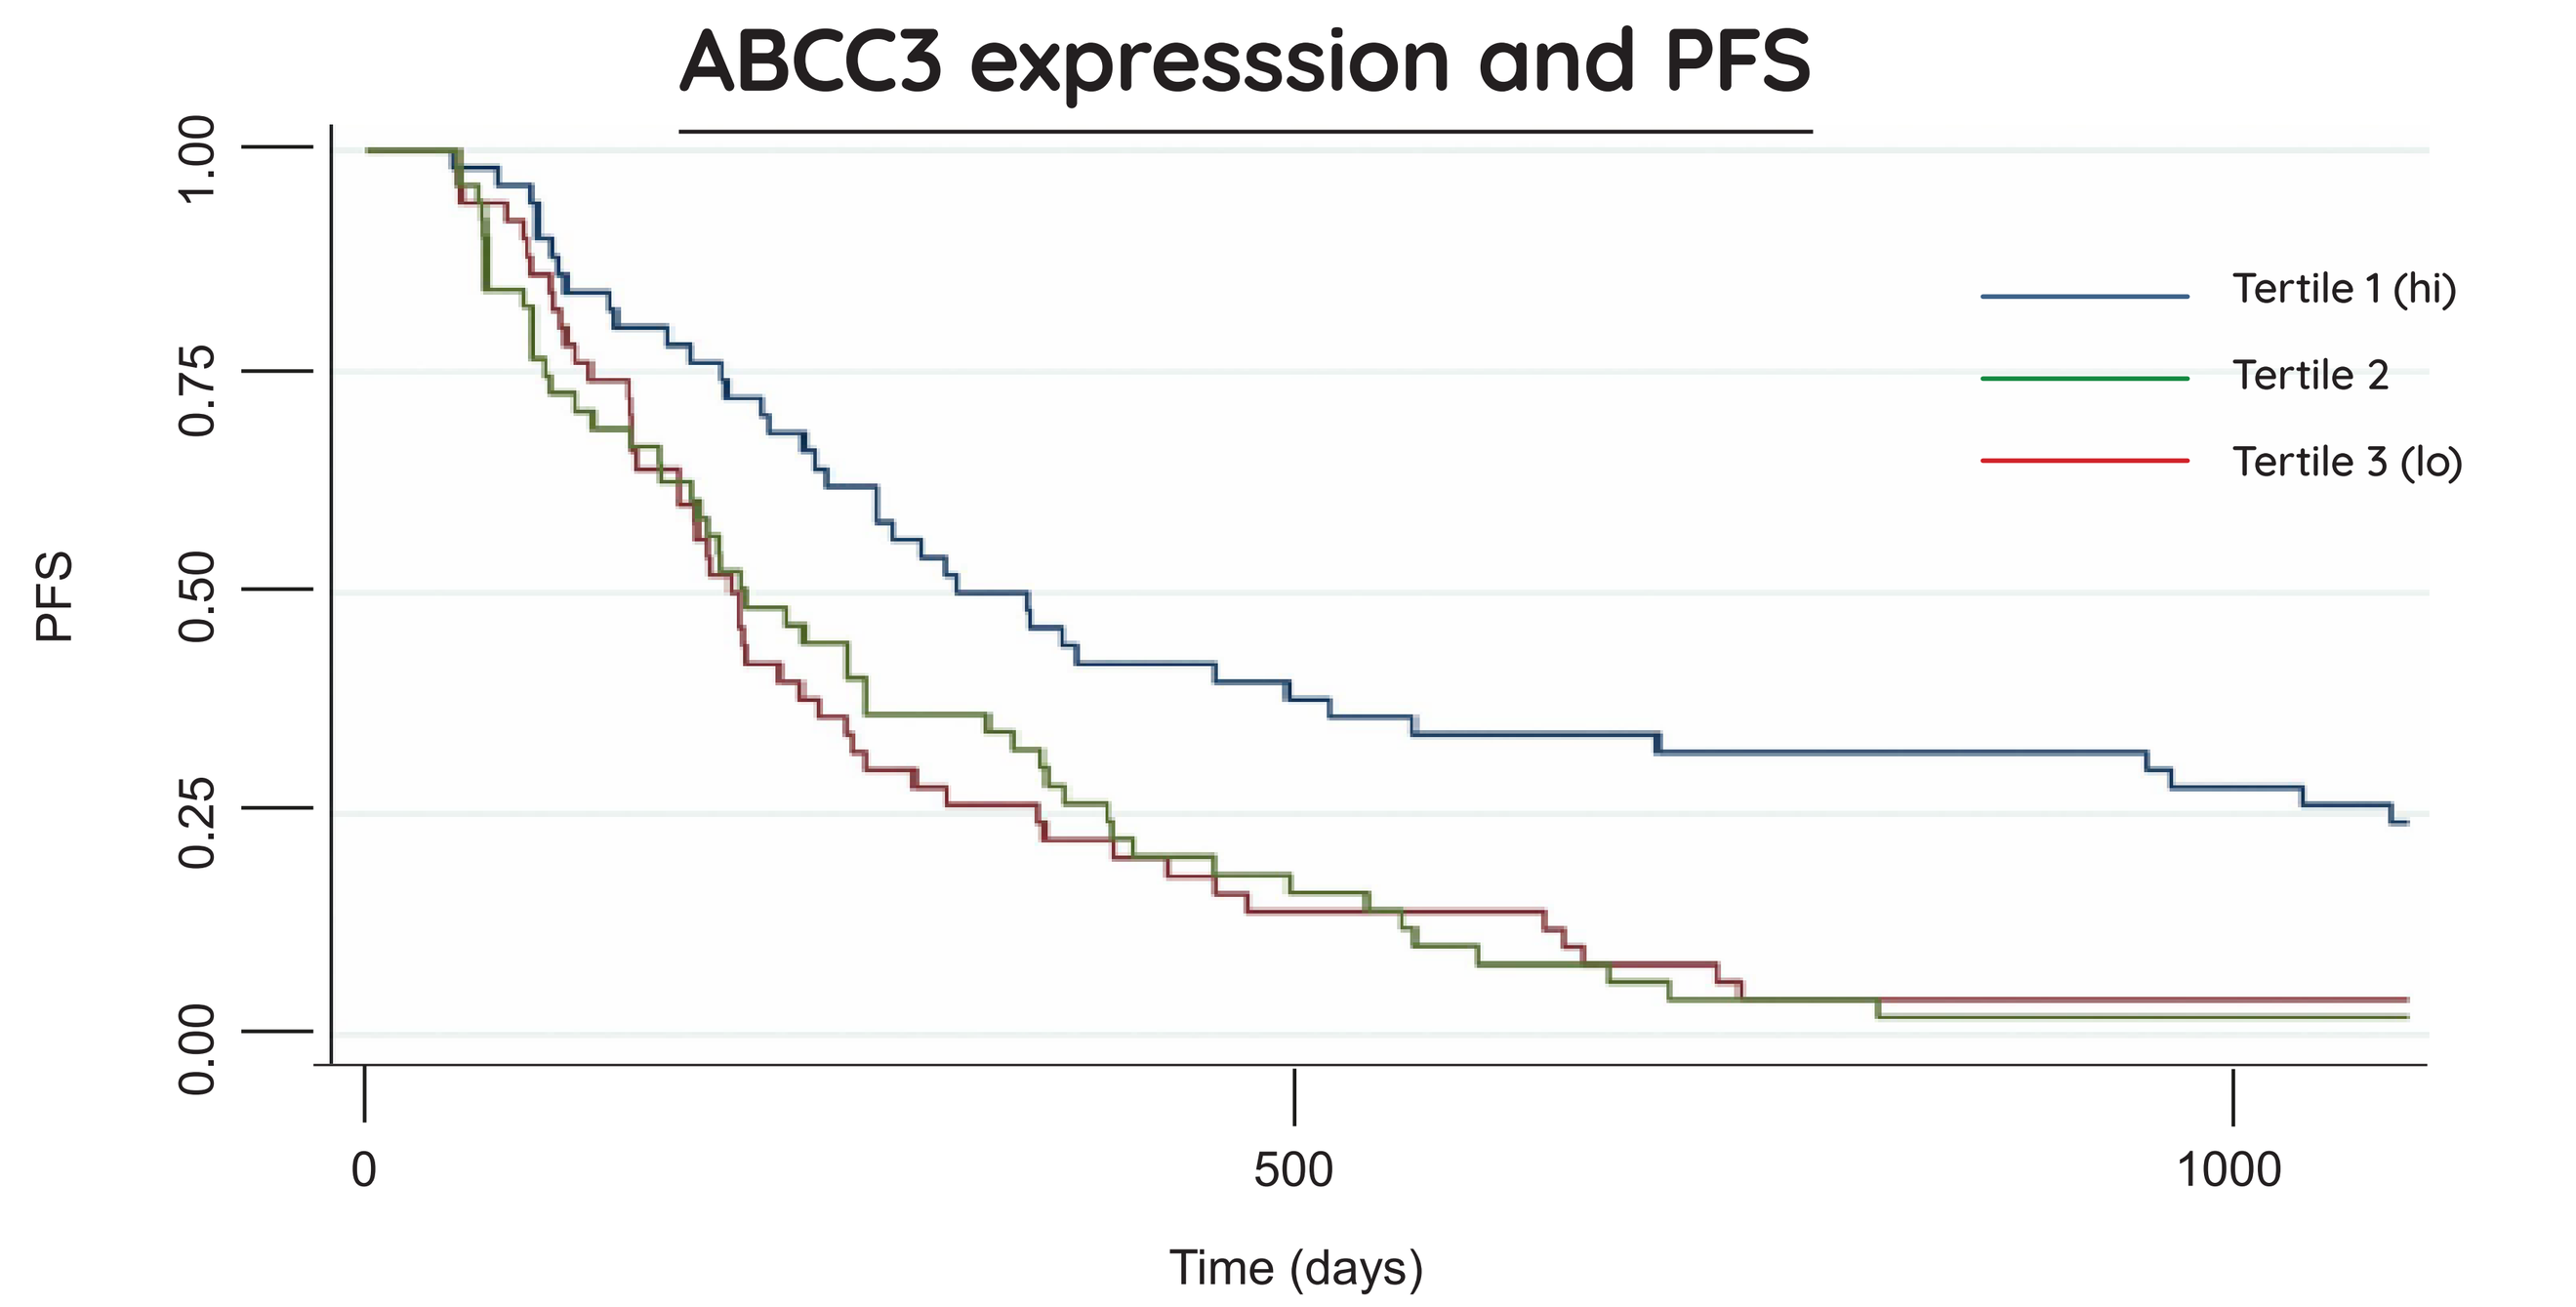

ABCC3 expresssion and PFS
PFS
1.00
0.75
0.50
0.25
0.00
Tertile 1 (hi)
Tertile 2
Tertile 3 (lo)
0
500
1000
Time (days)

Figure 1. Kaplan-Meier Curves showing PFS when patients with synchronous mCRC were categorised based on ABCC3 expression (Tertile 1/high expression (blue; n=50): 3.829 – 5.685, Tertile 2/intermediate expression (green; n=50): 5.695 – 6.461, Tertile 3/low expression (red; n=51): 6.513 – 9.009).

CONCLUSIONS

Folate-associated genes may predict response to treatment with FLV in stages III & IV CRC[2]. The similar overall prognosis of the patients in the lowermost and medium tertiles is perhaps part of the explanation for the low over-all response rate after treatment with FLV. It is interesting that the contribution of ABCC3 expression is independent of ECOG grade and other clinical variables.

Direct treatment with the active LV-metabolite is warranted to support the causal importance of metabolic activation of LV.

REFERENCES & ACKNOWLEDGEMENTS

References: **1.** Danenberg PV, Gustavsson B, Johnston P, Lindberg P, Moser R, Odin E, Peters GJ, Petrelli N (2016). Folates as adjuvants to anticancer agents: Chemical rationale and mechanism of action. Crit Rev Oncol Hematol 106:118-31. **2.** Odin E, Sondén A, Gustavsson B, Carlsson G, Wettergren Y (2015). Expression of Folate Pathway Genes in Stage III Colorectal Cancer Correlates with Risk of Recurrence Status Following Adjuvant Bolus 5-FU-based Chemotherapy. Mol Med 21: 597-604

Acknowledgements: We thank H. Björkqvist and A-L. Helminen for collection of surgical samples, L. Munro for work with the clinical database, and J. Flach and M. Åkerström for technical assistance. We also thank personnel at TATAA Biocenter for performing gene expression analyses. This work was supported by grants from the Swedish Cancer Society, the King Gustav V Jubilee Clinic Foundation for Cancer Research, the Swedish state under the LUA/ALF agreement, the IngaBritt and Arne Lundberg Foundation, and Isofol Medical AB.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at ASCO 2018, Poster no.(Board) 3550 (#43) Contact person: Prof. Bengt Gustavsson, e-mail: bengt.gustavsson@vgregion.se